Baruch S. Blumberg Institute — Est. by the Hepatitis B Foundation

Leronlimab, a humanized monoclonal antibody to CCR5, blocks breast cancer metastasis and enhances cell death induced by DNA damaging chemotherapy

Xuanmao Jiao[1], Min Wang[1], Zhiping Li[1], Richard G. Pestell[1, 2, 3, *]

[1]Pennsylvania Cancer and Regenerative Medicine Research Center, Baruch S. Blumberg Institute, 3805 Old Easton Road, Doylestown, PA., 18902.

[2]CytoDyn Inc., 1111 Main St, Suite 660, Vancouver, WA, 98660. [3]Lee Kong Chian School of Medicine, Nanyang Technological University, 637551, Singapore.

Abstract

Purpose of the study. To assess binding and functional interaction of the humanized monoclonal antibody to CCR5 (Leronlimab) with human breast cancer cell lines. The G protein coupled receptor CCR5, is normally expressed on a subset of T cells and serves as a co-receptor for HIV infection. During malignant transformation CCR5 expression is known to increase in a number of cancers (breast cancer (BCa), prostate cancer, colon cancer, melanoma). CCR5 targeted cancer clinical trials using small molecular inhibitors opened to accrual in late 2018. CCR5 is expressed in >50% of human BCa, primarily in triple negative BCa. Its expression in human BCa correlates with poor outcome and CCR5$^+$ BCa epithelial cells have characteristics of cancer stem cells, forming mammospheres and initiating tumors with >60-fold greater efficiency in mice. Reintroduction of CCR5 expression into CCR5 negative BCa cells promotes tumor metastases and induces DNA repair gene expression and activity. The CCR5 inhibitor Leronlimab has been used for treatment of >660 patients with HI, including meeting its primary endpoints in a phase III study, without significant adverse events reported. **Results.** Leronlimab bound to CCR5 expressed in human breast cancer cell lines with 98% efficiency. Leronlimab abrogated CCL5 induced Ca^{+2} flux and blocked 3-d matrigel invasion of MDA-MB-231 cells. Leronlimab blocks human breast cancer xenograft metastasis in mice. Leronlimab also augmented cell killing by DNA damage inducing agents including Doxorubicin. **Conclusions.** Leronlimab binds CCR5 in BCa cells, blocking breast cancer cellular invasion and tumor metastasis, and augmenting cell killing by DNA damage inducing chemotherapies. As CCR5 augments DNA repair and is expressed selectively on cancerous but not normal breast epithelial cells, Leronlimab may enhance the tumor specific activities of DDR-based treatments, allowing a reduction in dose of chemotherapy and radiation.

1.The binding of Leronlimab with CCR5 expressed in breast cancer cells

A

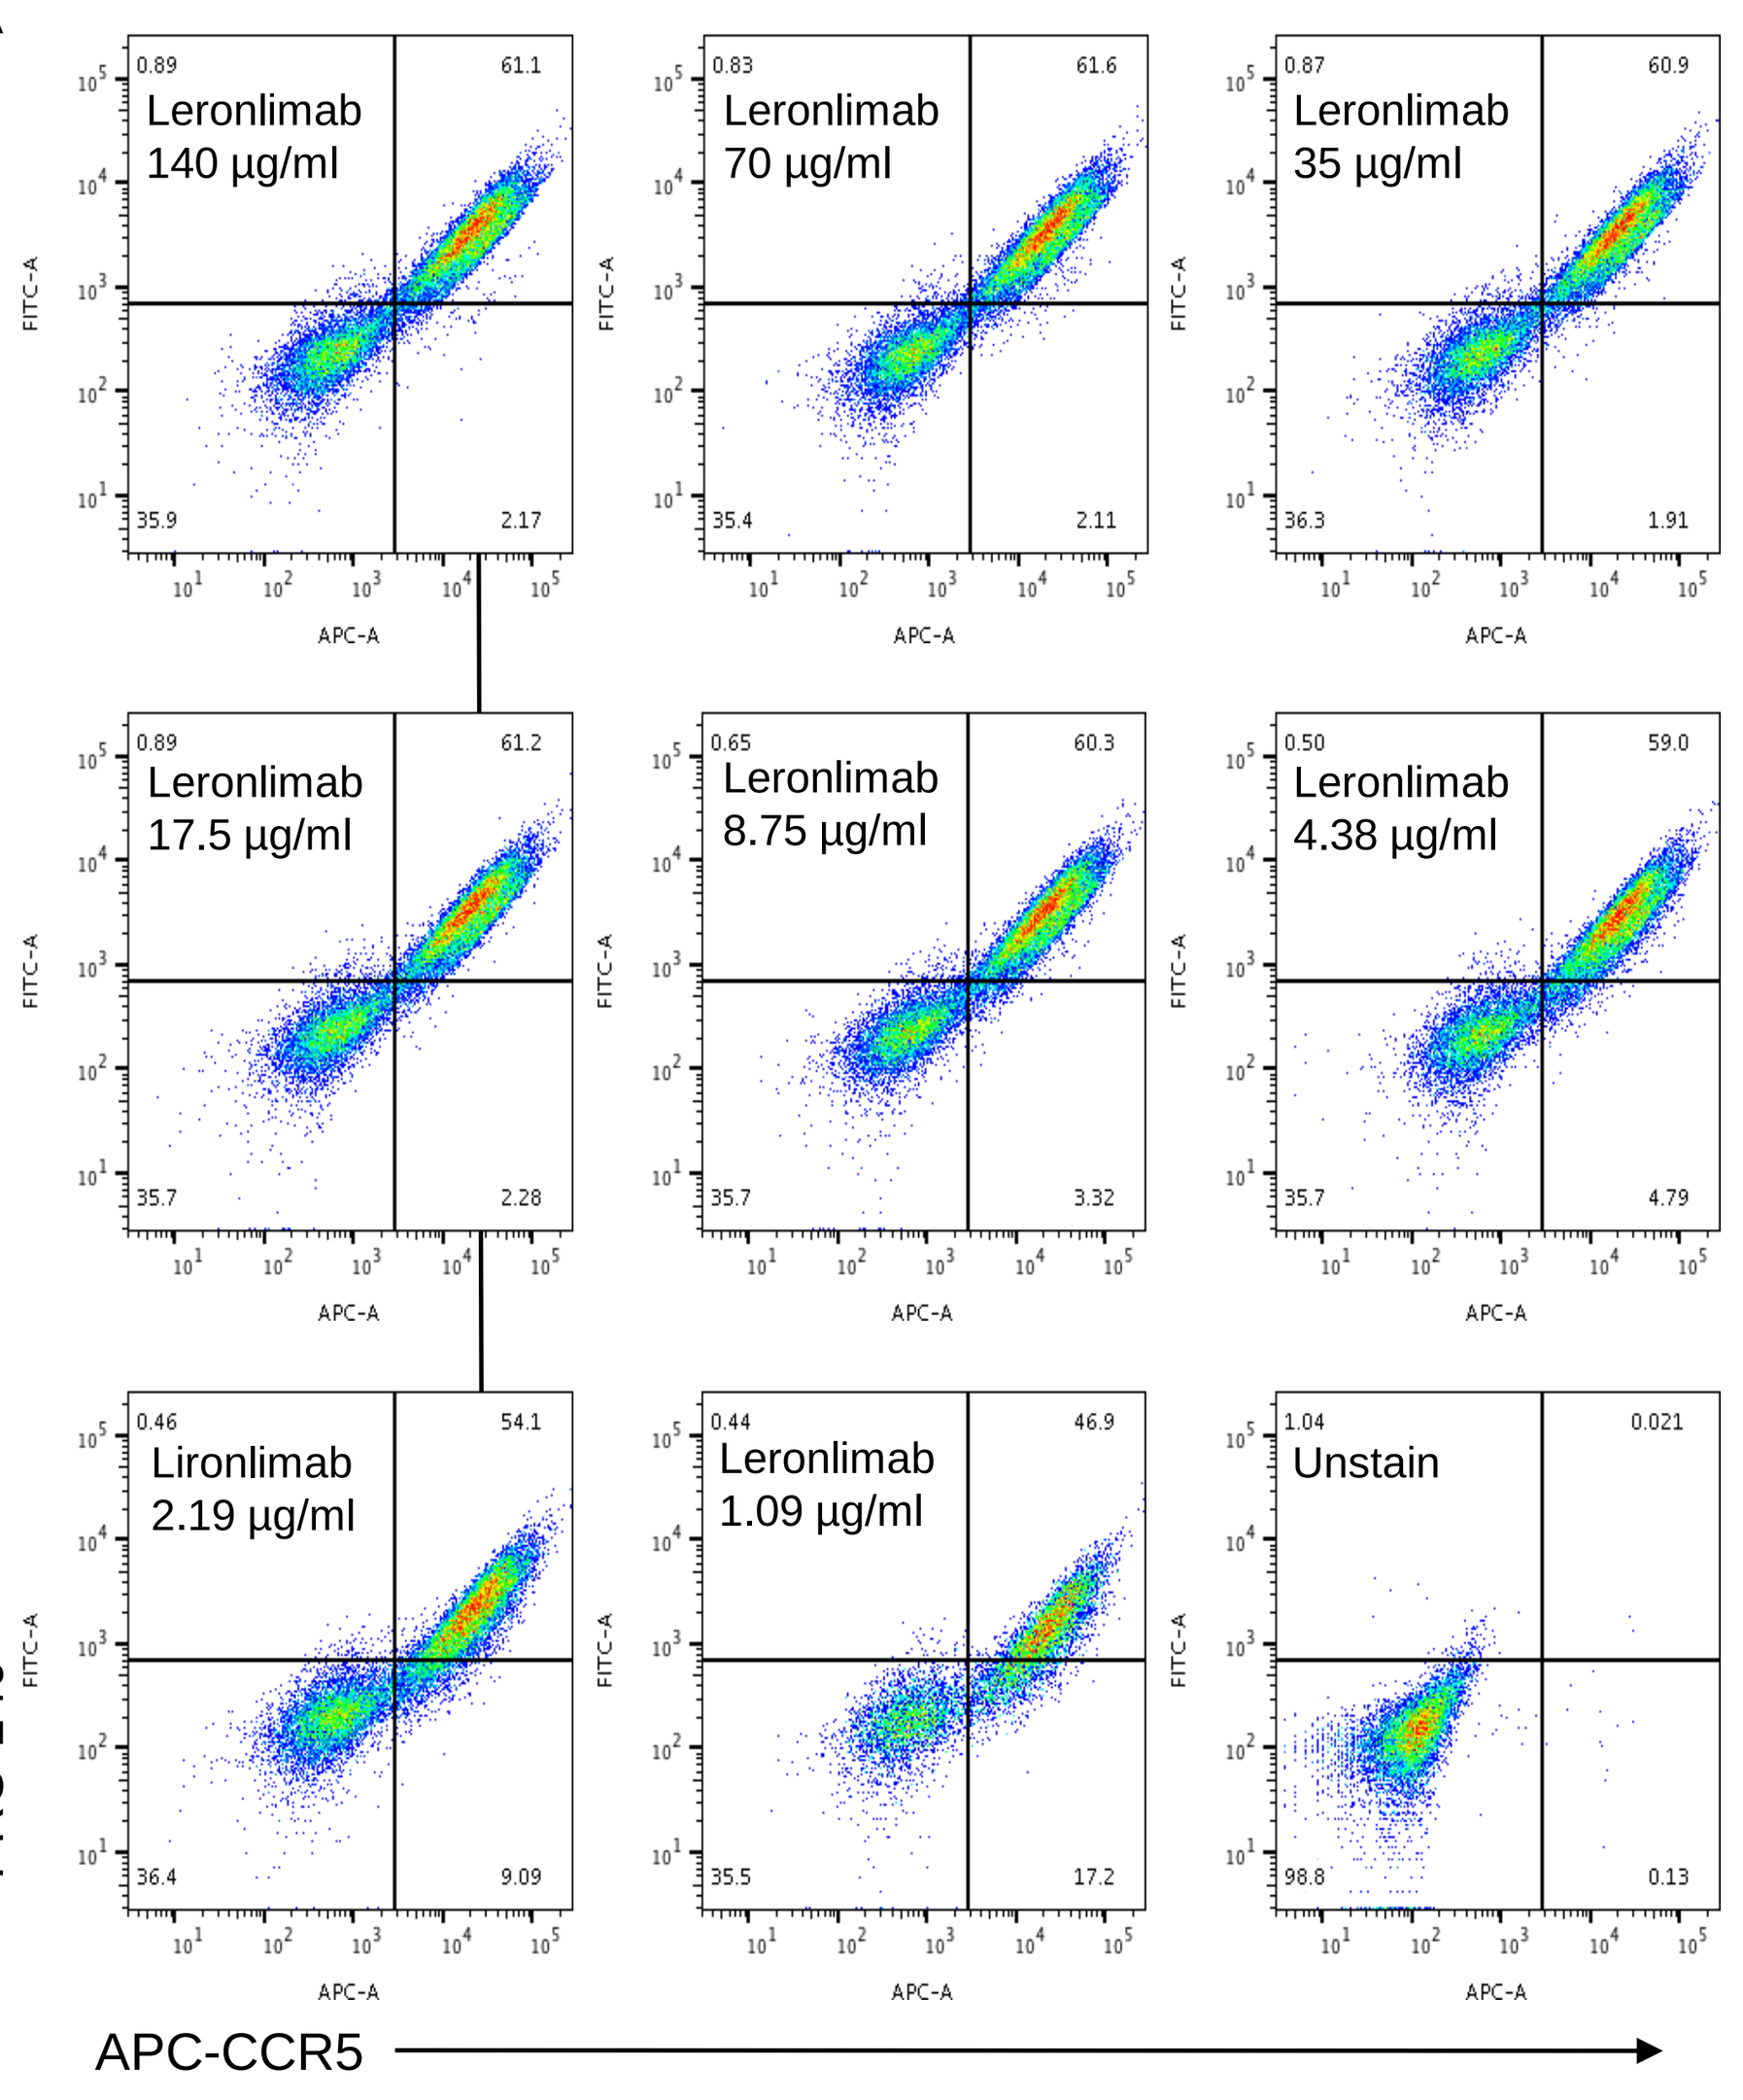

Leronlimab 140 µg/ml
Leronlimab 70 µg/ml
Leronlimab 35 µg/ml
Leronlimab 17.5 µg/ml
Leronlimab 8.75 µg/ml
Leronlimab 4.38 µg/ml
Lironlimab 2.19 µg/ml
Leronlimab 1.09 µg/ml
Unstain
PRO-140
APC-CCR5

B

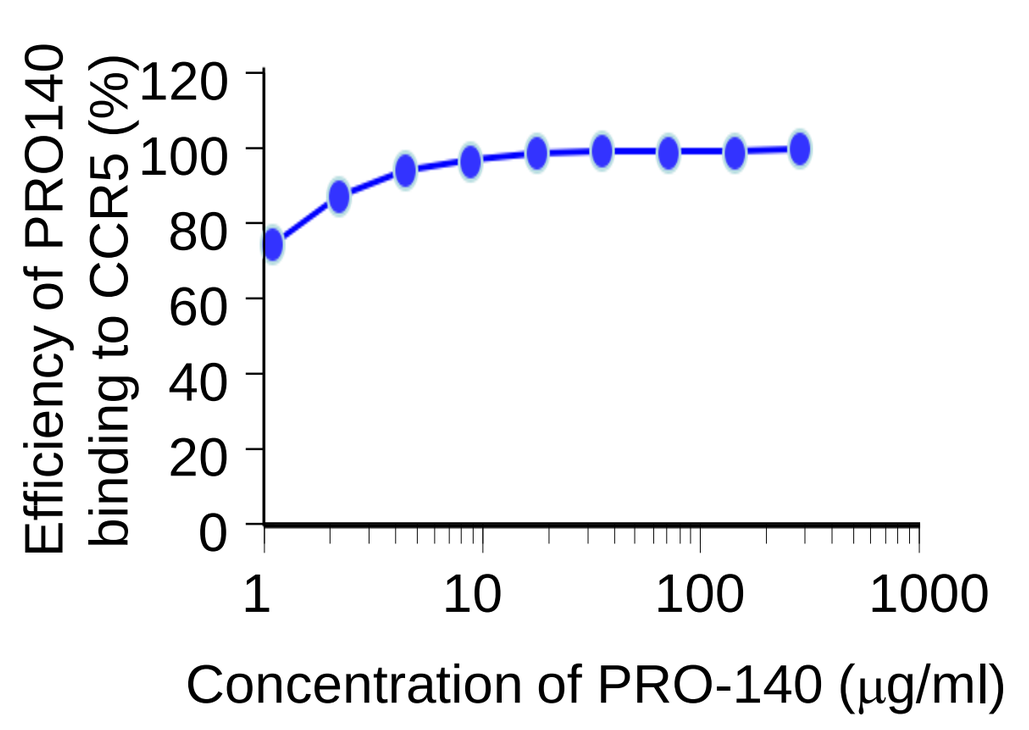

Efficiency of PRO140 binding to CCR5 (%)
Concentration of PRO-140 (µg/ml)

Figure 1. **Leronlimib binds CCR5 in human breast cancer cells**. (A). In order to determine the binding of Leronlimab to human CCR5 in breast cancer cells, we used an MDA-MB-231 human breast cancer cell line transfected with a human CCR5 expression vector as a model system. A commercial APC conjugated mouse anti-human/mouse/ rat CCR5 antibody from R&D (FAB1802A) (APC-αCCR5) which we had previously tested was used as a positive control to assess CCR5 positive cells. MDA-MB-231-CCR5 cells were stained with both APC-αCCR5 and Leronlimab using the concentration from 1-140 µg/ml. Alexa Fluor 488 conjugated mouse anti-human IgG was used as secondary antibody to measure Leronlimab binding cells. Analysis of Leronlimab binding with CCR5 by FACS is shown in (A). Leronlimab binding with human CCR5 was validated. (B).The efficiency of PRO140 binding to CCR5 positive cells was up to 98%

2. The effects of PRO140 on CCL5 induced Ca^{2+} responses in MDA-MB-231-CCR5 cells

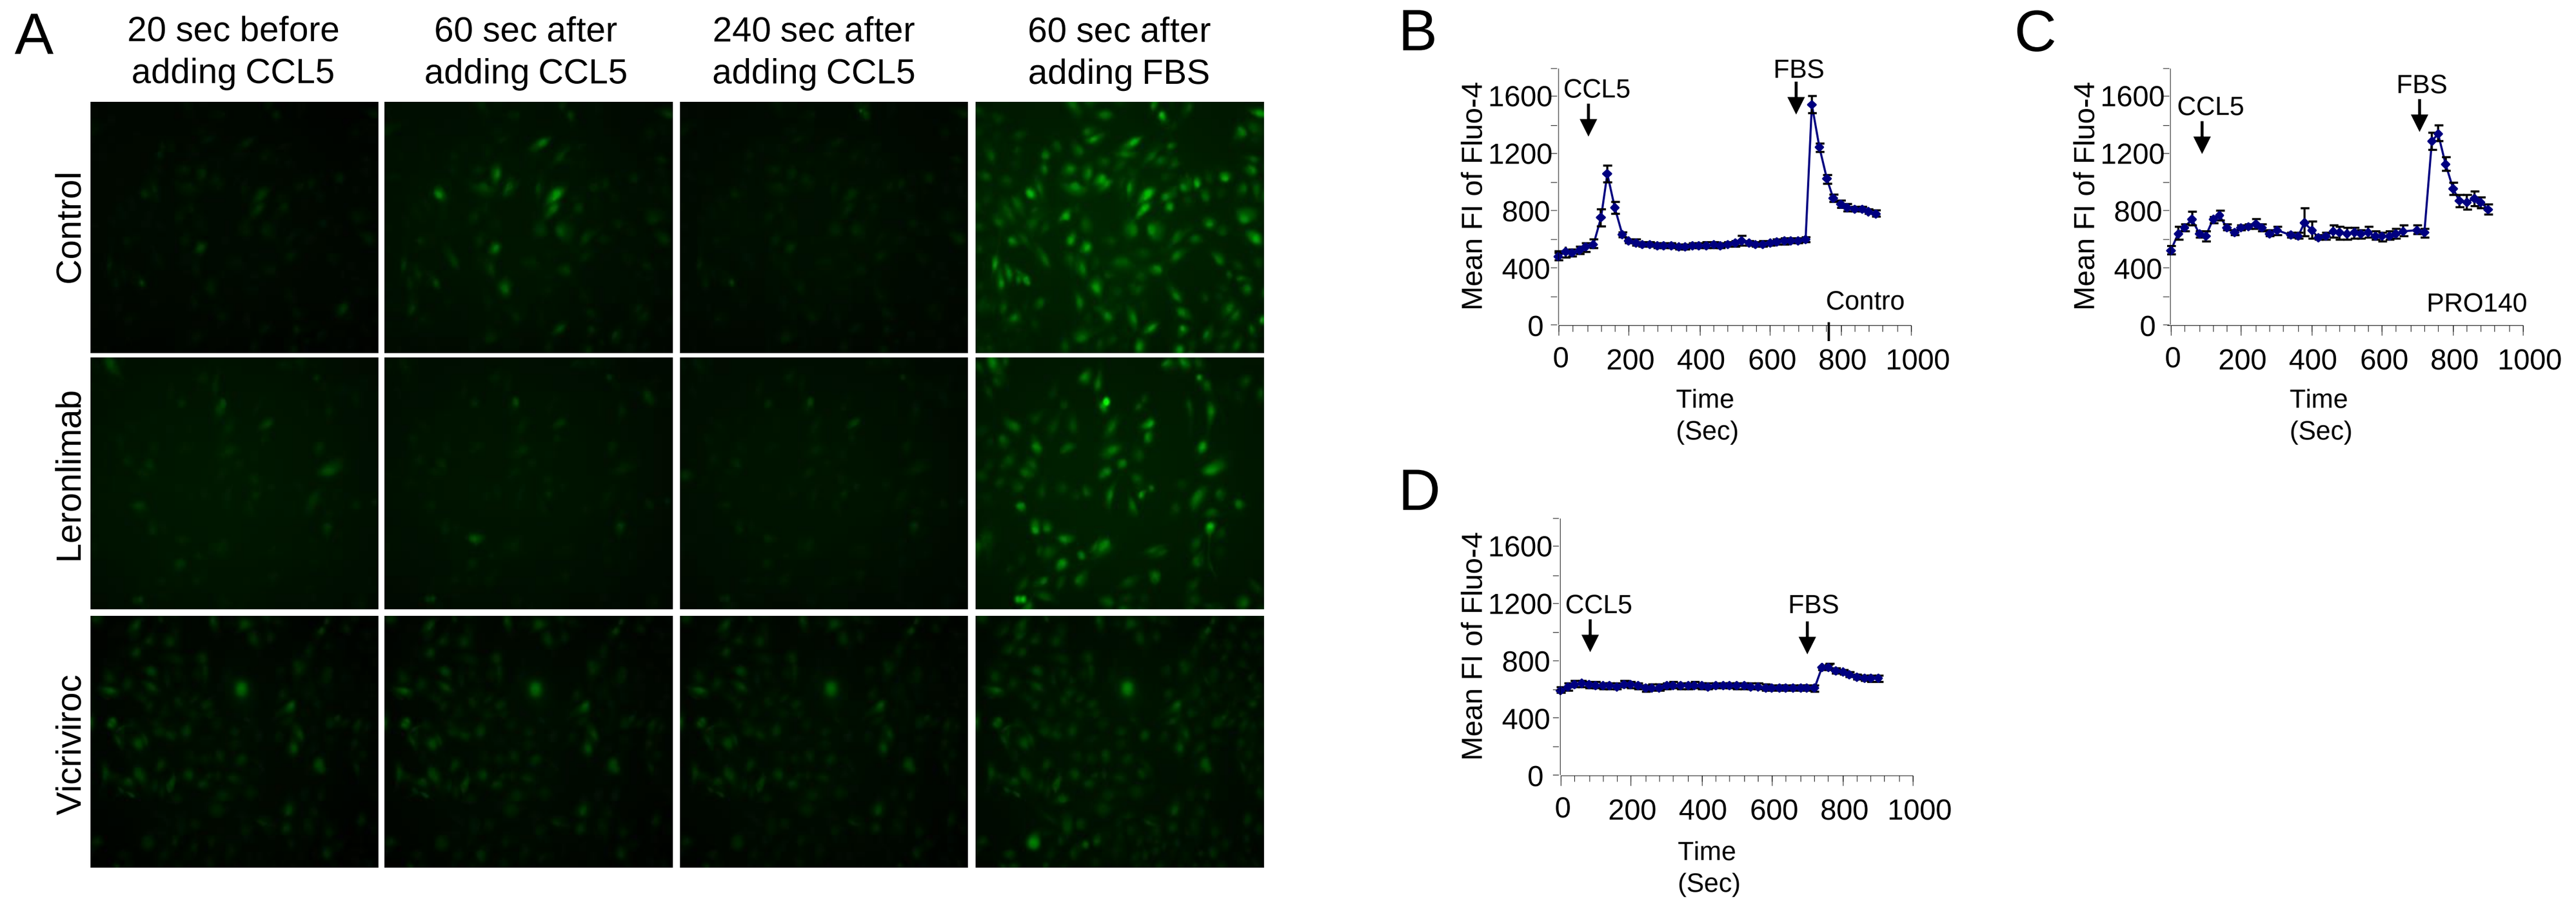

A
20 sec before adding CCL5
60 sec after adding CCL5
240 sec after adding CCL5
60 sec after adding FBS
Control
Leronlimab
Vicriviroc
B
C
D
CCL5
FBS
Mean FI of Fluo-4
Time (Sec)
Contro
PRO140

Figure 2. **PRO140 blocks human CCR5-mediated signaling in human breast cancer cells**. CCR5 activation induces calcium flux (Mueller et al., 2002; Petkovic et al., 2004). To assess the effects of Leronlimab on CCRR5 function, we measured the calcium responses induced by CCL5 in MDA-MB-231-CCR5 cells with or without Leronlimab by living cell image (Figure 2A-C). Fluo-4 was used as calcium concentration indicator. The CCR5 antagonist, Vicriviroc, was used as positive control (Figure 2A, D). The results showed that Leronlimab can block CCL5 induced calcium responses in MDA-MB-231-CCR5 cells (1.23±0.10, N=10 for control cells and 0.54±0.13 N=12 for PRO140 treated cells. P<0.001 at calcium peak induce by CCL5).

3. Leronlimab blocks breast cancer cell 3D-matrigel invasion

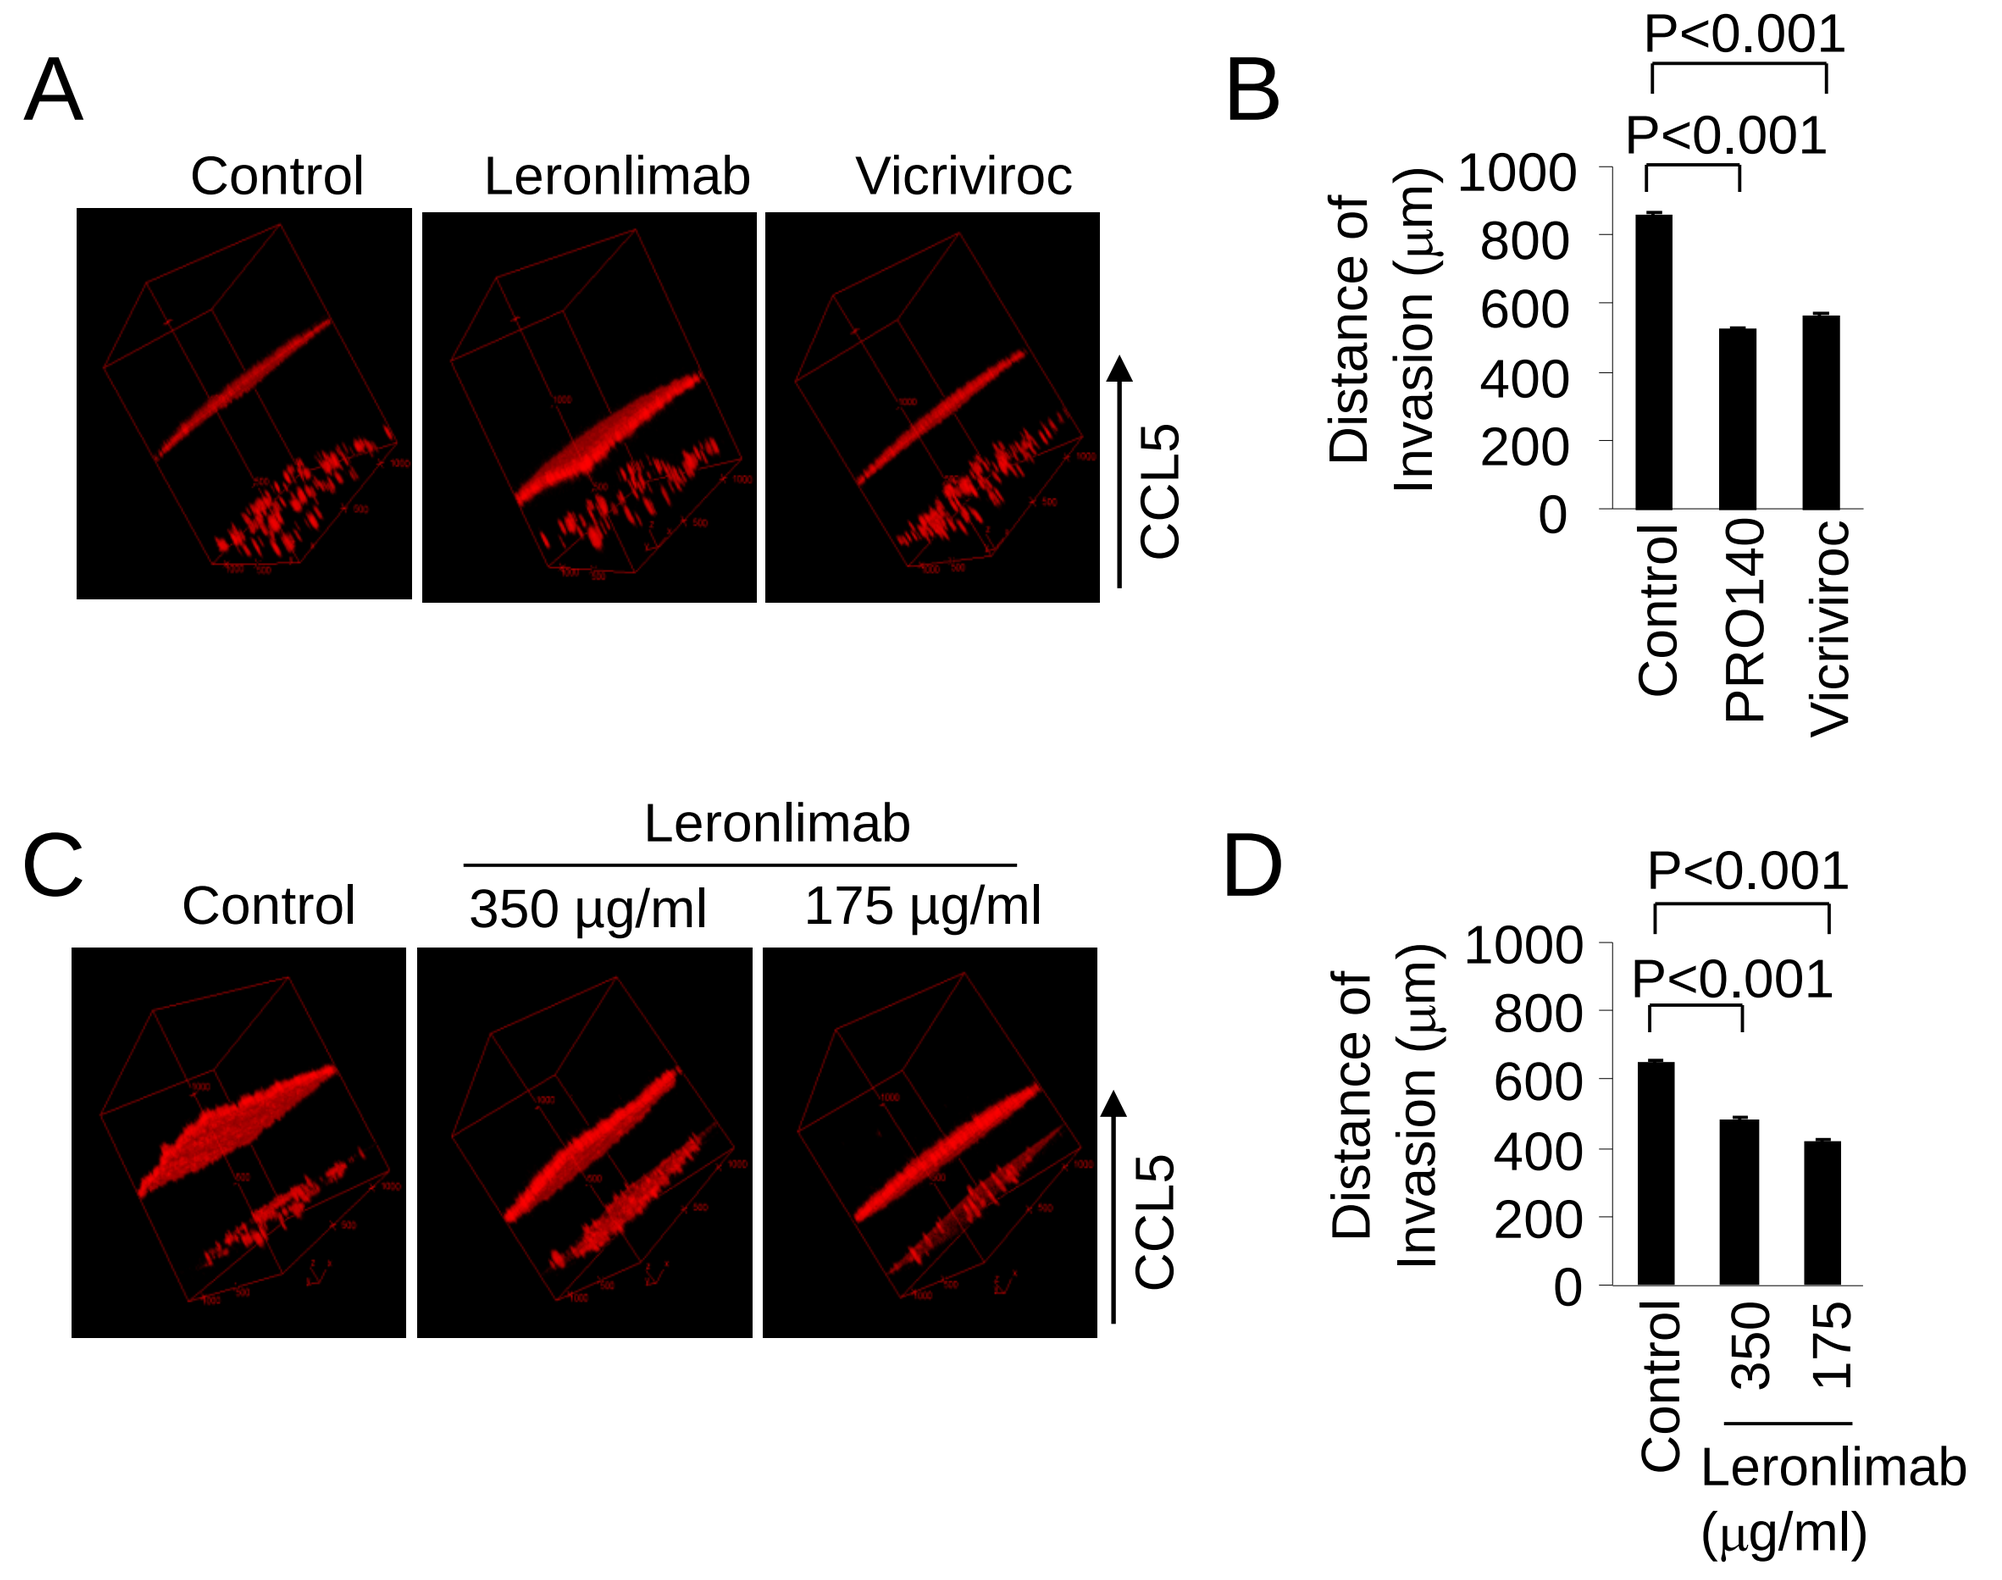

A
Control
Leronlimab
Vicriviroc
CCL5
B
P<0.001
P<0.001
Distance of Invasion (µm)
Control
PRO140
Vicriviroc
C
Leronlimab
Control
350 µg/ml
175 µg/ml
CCL5
D
P<0.001
P<0.001
Distance of Invasion (µm)
Control
350
175
Leronlimab (µg/ml)

Figure 3. **Leronlimab blocks CCR5 mediated invasion of human breast cancer cells into extracellular matrix.** The ability of breast cancer cells to invade extra-cellular matrix is distinguishable from but an important step in tumor metastasis (Zetter, 1990). To test the ability of PRO140 to block 3D-matrigel invasion assay, MDA-MB-231 cells were used. CCL5 was used as chemoattractant to induce invasion. The small molecule inhibitor of CCR5, Vicriviroc, was used as a form of positive control. Leronlimab reduced CCL5-induced MDA-MB-231 breast cancer cell invasion with similar efficacy as Vicriviroc (A, B) (855±9, N=8 for control vs 855±9, N=9 for Leronlimab, P <0.001). We also tested the effects of different dose of Leronlimab on breast cancer cell invasion and the results showed that both 175 and 350 µg/ml of Leronlimab can effectively block MDA-MB-231 cell invasion (C, D).

4. Leronlimab blocks breast cancer cell metastasis in a mouse lung metastasis model

A

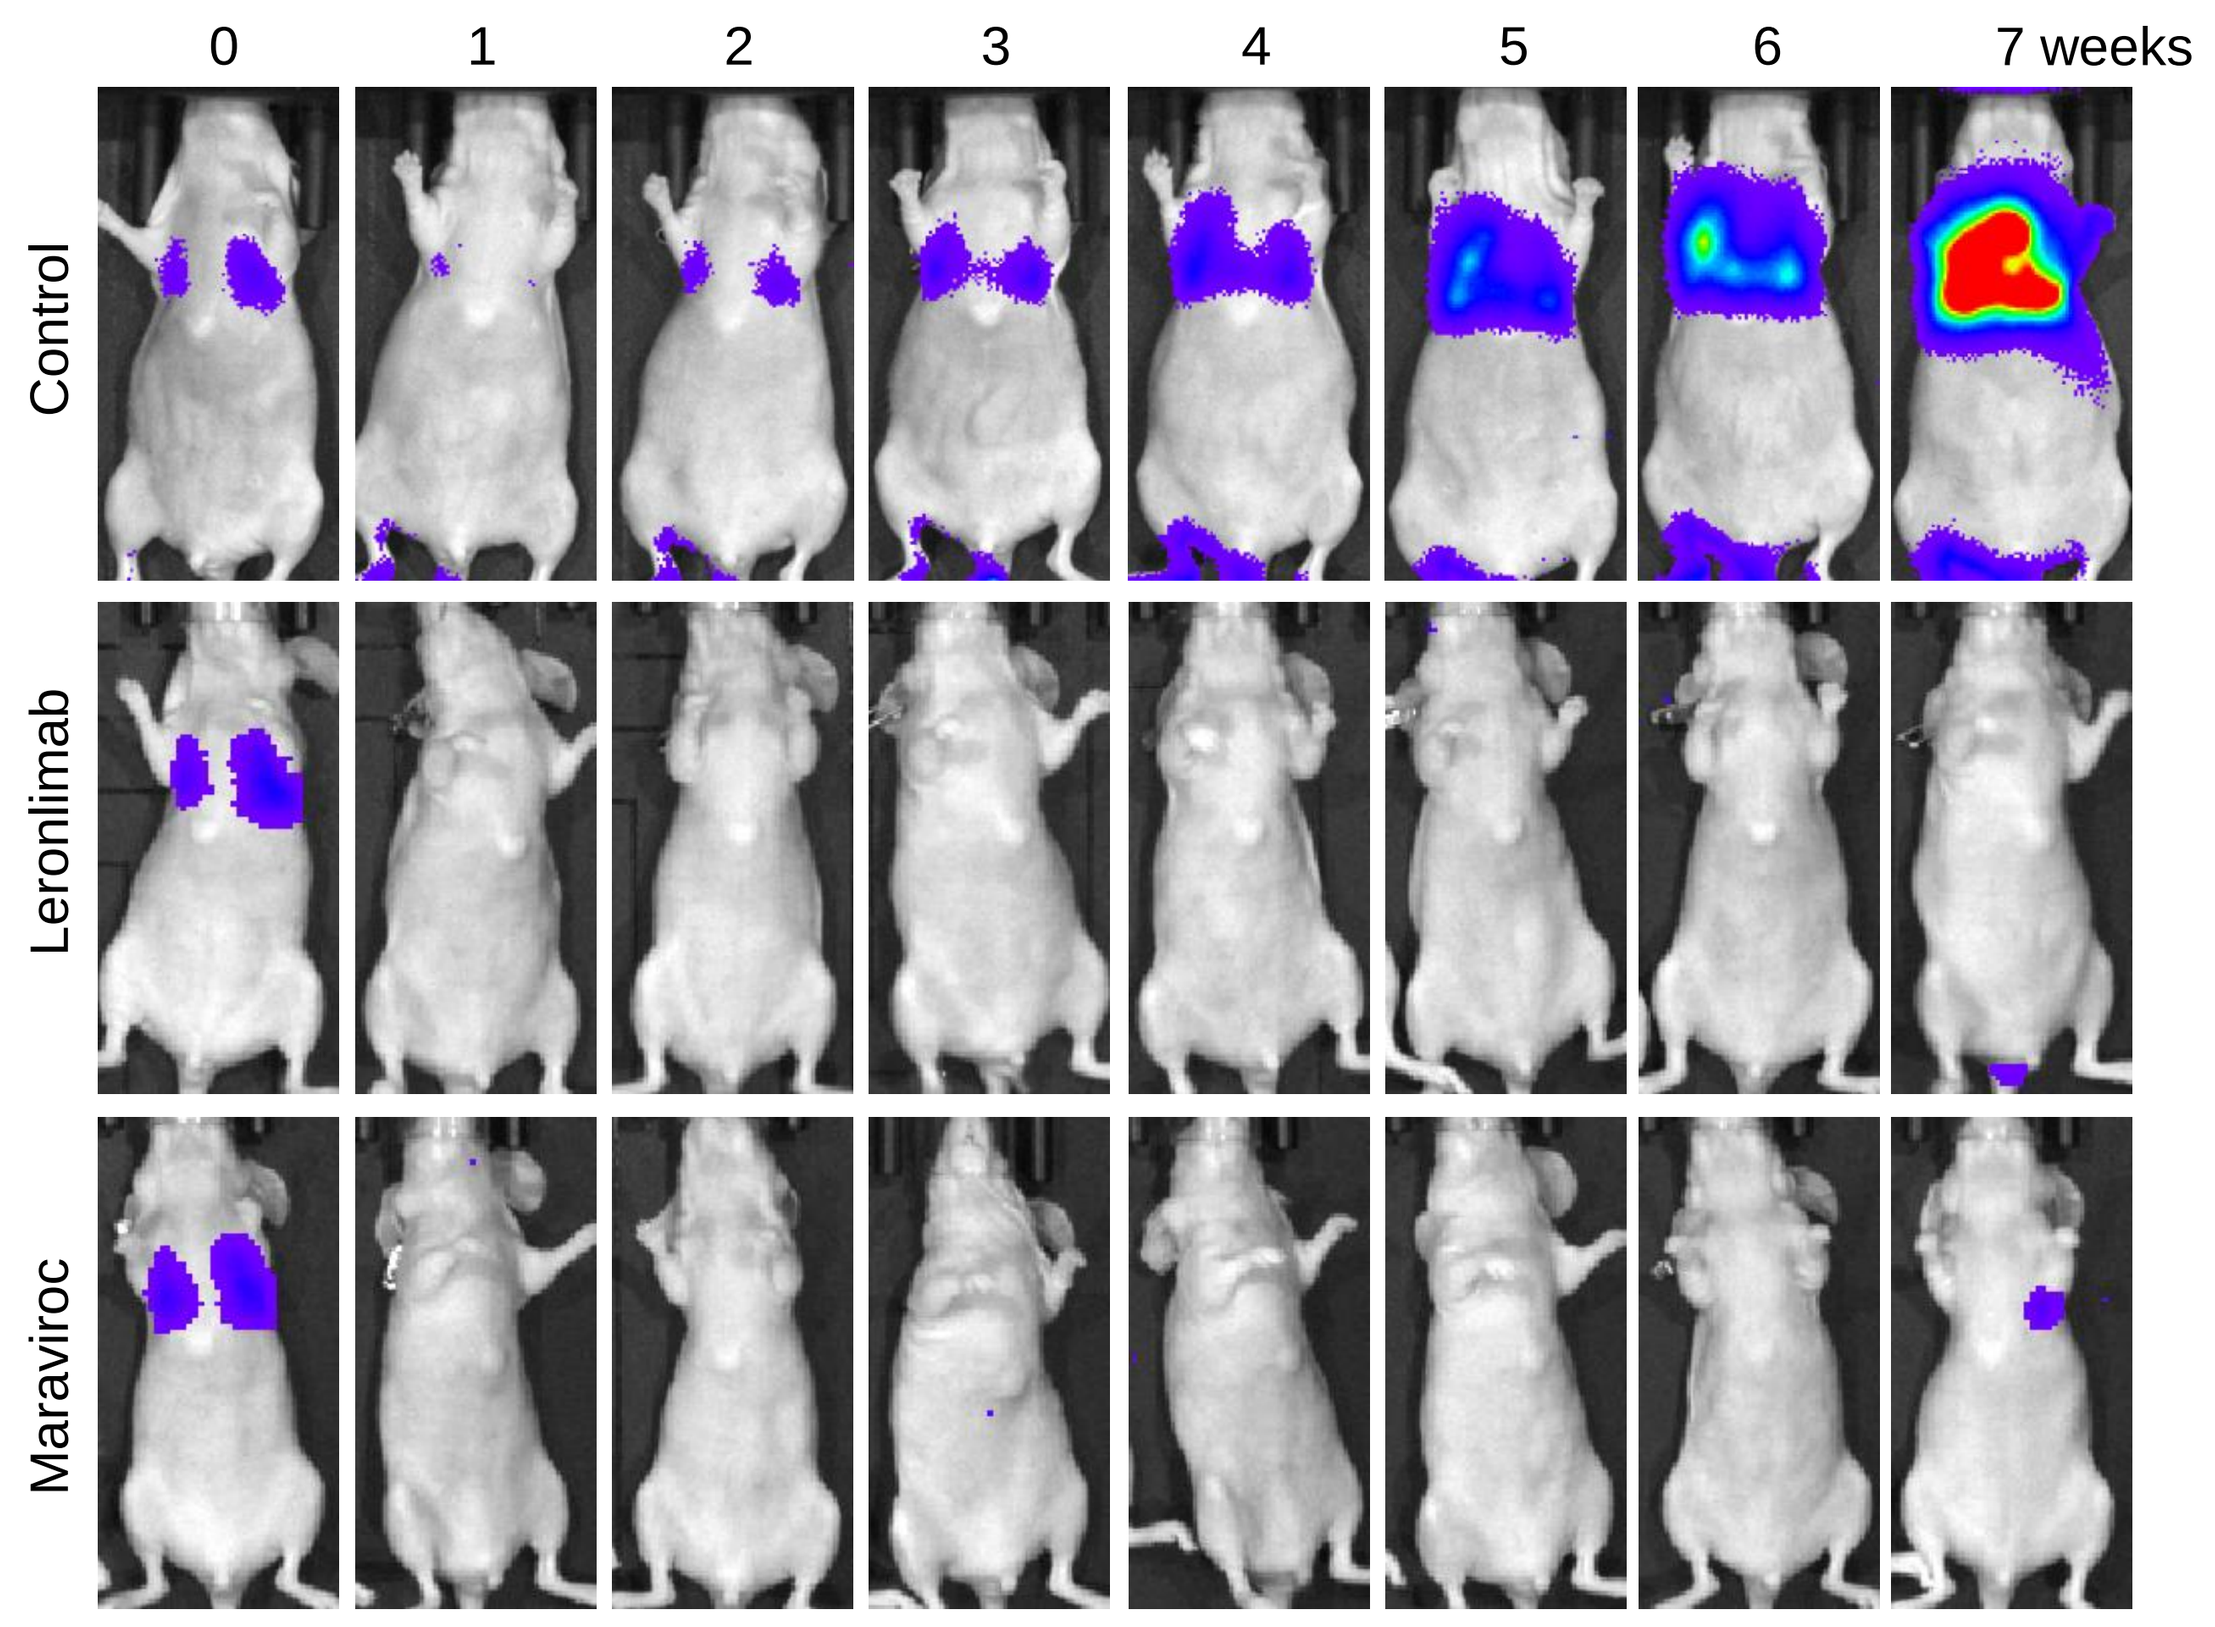

0
1
2
3
4
5
6
7 weeks
Control
Leronlimab
Maraviroc

B

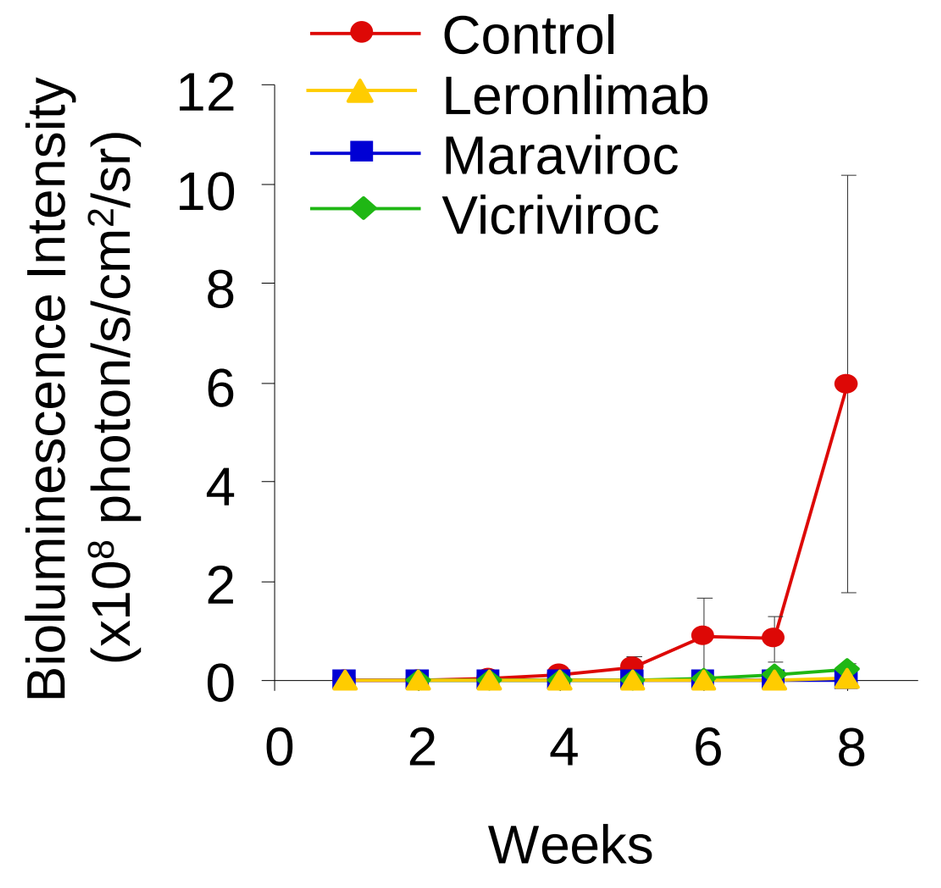

Control
Leronlimab
Maraviroc
Vicriviroc
Bioluminescence Intensity (x10⁸ photon/s/cm²/sr)
Weeks

Figure 4. Leronlimab block breast cancer metastasis in mice. The mice were divided into 4 groups (control, Leronlimab, Maraviroc and vicriviroc) randomly. MDA-MB-231 cells stable transfected with Luc2-GFP was injected into the mice through tail-vein. The mice in each group were treated one day before injection. The metastasis tumor formed in the lung was determined by bioluminescence imaging. The bioluminescence images of the representative mice from control, Leronlimab and Maraviroc group were showed in (A). The quantitative analysis of tumor size in each group was showd in (B). The size of tumors defined by photon flux (x10^8 p/sec/cm^2/sr). The data was showed as Mean ± SE. Leronlimab dramatically decreased breast cancer tumor metastais to the lung.

5. *Leronlimab enhances cell death induced by Doxorubicin*

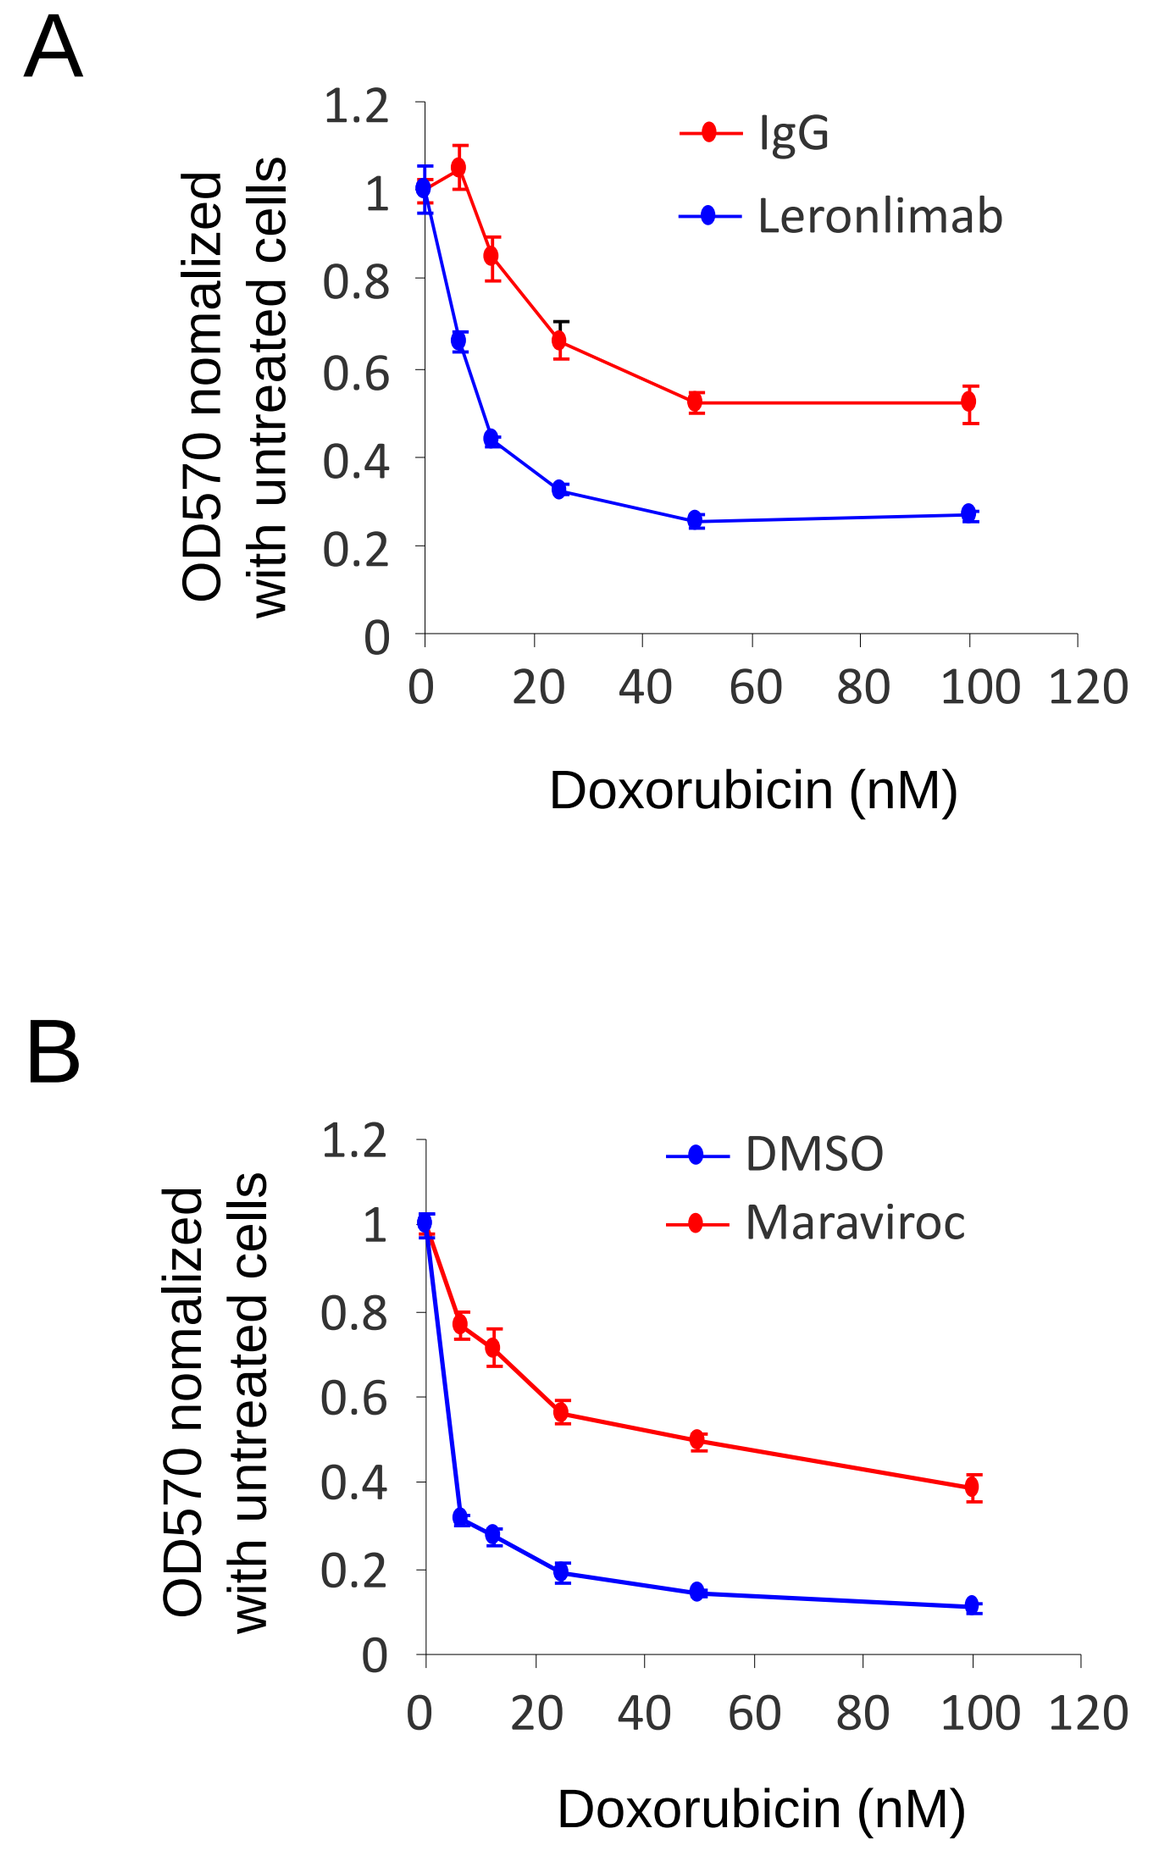

A
IgG
Leronlimab
OD570 nomalized with untreated cells
Doxorubicin (nM)
B
DMSO
Maraviroc
OD570 nomalized with untreated cells
Doxorubicin (nM)

Figure 5. Leronlimab enhances the cell death induced by Doxorubicin, a DNA damage inducing chemotherapy agent. MDA-MB-231 cells was treated with 10 mg/ml of Leronlimab combining with different dose of Doxorubicin for 3 days. The MTT assay was used to determine the relative cell number (A). The cells treated with Maraviroc (100 mM) combining with different dose of Doxorubicin were used as positive control (B). Data are shown as mean ± SEM for N= 8.